UNIVERSITY OF ROCHESTER **EYE INSTITUTE**

ARVO 2009
Program # : 4647
Poster #: D891

Extension of Tear Film Break-Up by Novel Formulation of Glycerin 1%

W.G. Gensheimer[1], D.M. Kleinman[2,3], M.O. Gonzalez[2], D. Sobti[1], A. Loxley[4], M. Mitchnick[4], J.V. Aquavella[2]
[1]School of Medicine and Dentistry, University of Rochester, Rochester, NY; [2]University of Rochester Eye Institute, Rochester, NY;
[3]Eyeon Therapeutics, Inc., Rochester, NY; [4]Particle Sciences, Inc., Bethlehem, PA

SCHOOL OF MEDICINE & DENTISTRY
UNIVERSITY of ROCHESTER MEDICAL CENTER

PURPOSE

One way to evaluate the effectiveness of an artificial tear preparation is to measure the tear film break up time (TFBUT) before and after eye drop instillation. Currently, even the best products on the market provide only limited extensions of TFBUT.[1-4] Furthermore, most artificial tears lose their effectiveness after roughly one hour.[1-4] Drug product formulation is critical to maximizing the benefits of an active pharmaceutical ingredient. A new formulation of glycerin 1% was compared to a market leading artificial tear product to determine if a specifically engineered formulation could significantly improve the activity of commonly used ophthalmic demulcents.

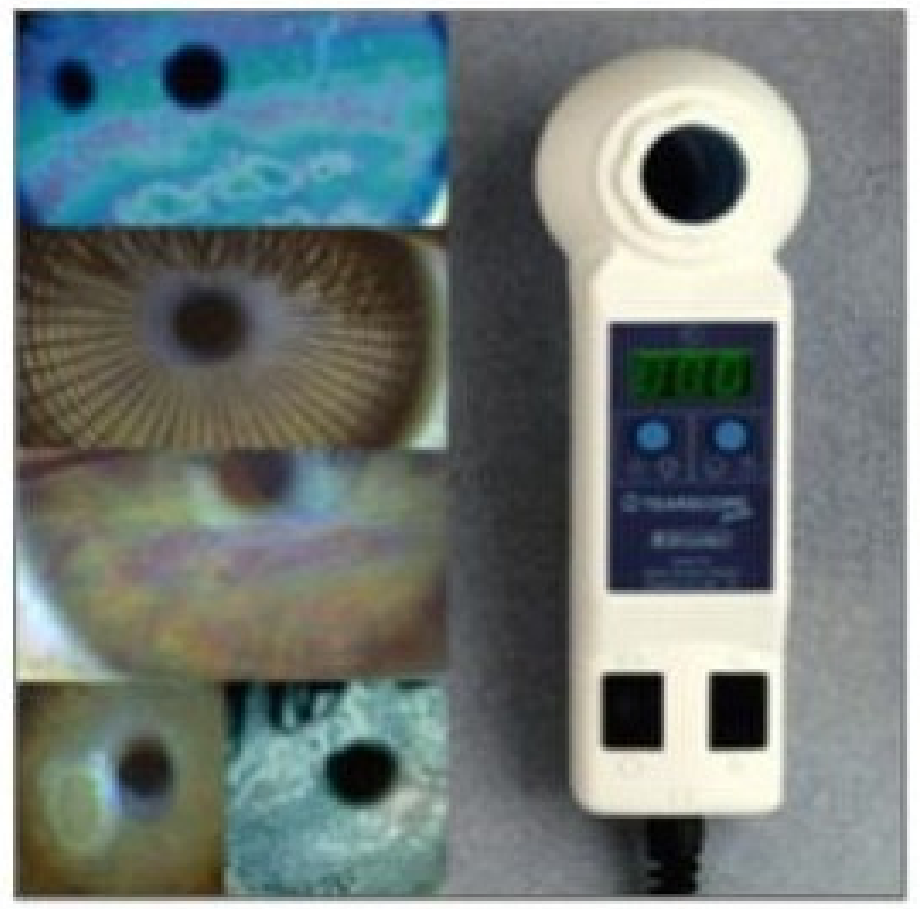

Image 1. Tearscope Plus™

Image downloaded from http://www.bon.de/images/tearscope.jpg

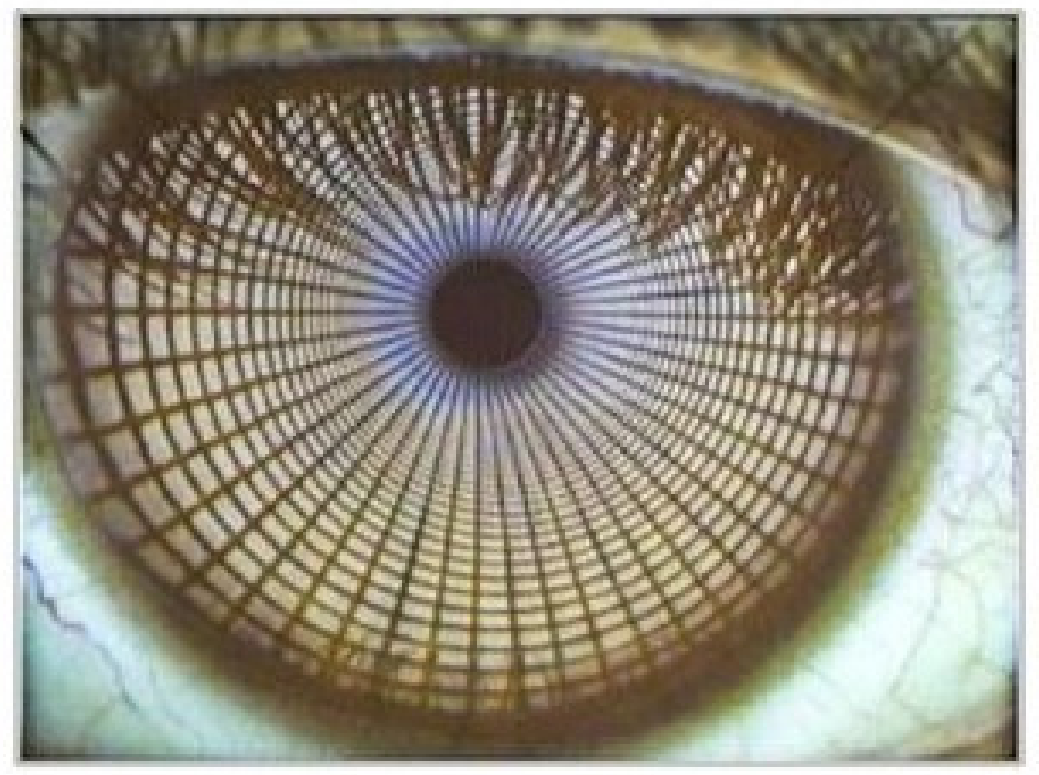

Image 2. Tearscope Plus™ (detail of corneal reflection used for NIBUT)

Image downloaded fromttp://www.bon.de/images/tearscope.jpg

METHODS

This single-center, single visit, randomized, double-masked exploratory trial compared the extension of TFBUT between a new formulation of glycerin 1% and a commercially available artificial tear formulation of propylene glycol (0.3%) and polyethylene glycol (0.4%). All subjects received a single instillation of 30 µL of each product randomly assigned to one of their eyes. Non-Invasive Break-Up Time (NIBUT) and Fluorescein Break-Up Time (FBUT) were measured in 3 groups of subjects (N=16) in order to assess the performance of these products in patients with different stages of dry eye disease. The study groups were asymptomatic to mild (Group 1; n=5), mild to moderate (Group 2; n=5), and moderate to severe (Group 3; n=6). NIBUT was measured using the Tearscope Plus (Image 1 and 2) at pre-instillation and again at 15, 30, 60, and 120 minutes after instillation. FBUT was measured at 120 minutes after instillation. The tenets of the Declaration of Helsinki were followed in this study. The informed consent and protocol were approved by the Research Subjects Review Board (RSRB) at the University of Rochester School of Medicine and Dentistry in Rochester, New York.

EYEON THERAPEUTICS

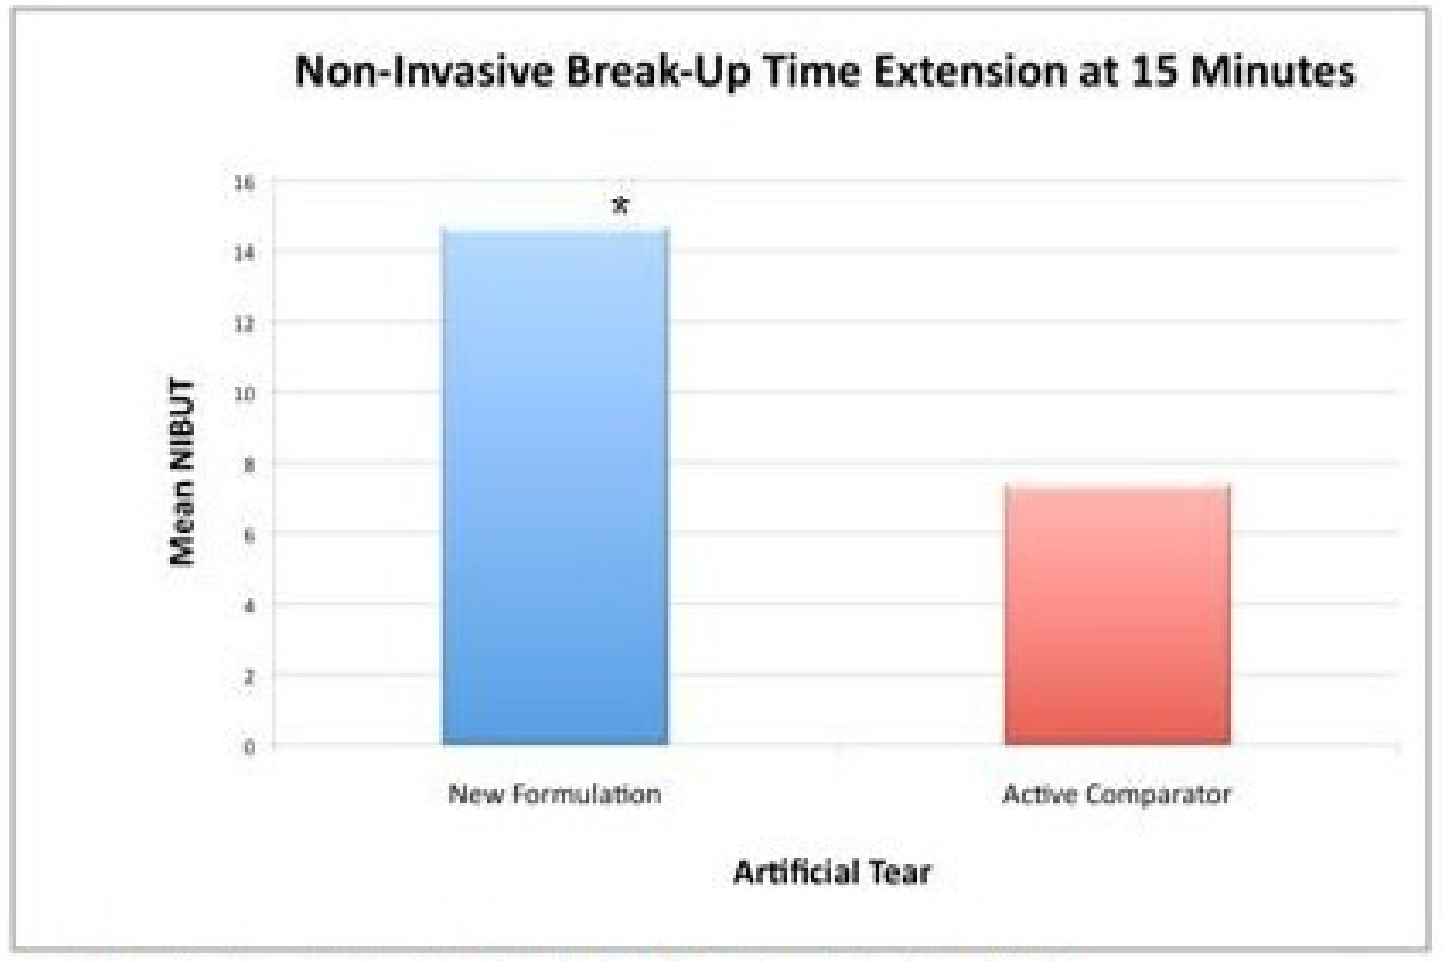
Non-Invasive Break-Up Time Extension at 15 Minutes
Mean NIBUT
0
2
4
6
8
10
12
14
16
*
New Formulation
Active Comparator
Artificial Tear

Figure 1. Mean Non-Invasive Break-Up Time (NIBUT) Extension at 15 minutes (N=16)
* The new formulation containing glycerin 1% significantly extended NIBUT by 14.67 seconds at 15 minutes (p = 0.05)

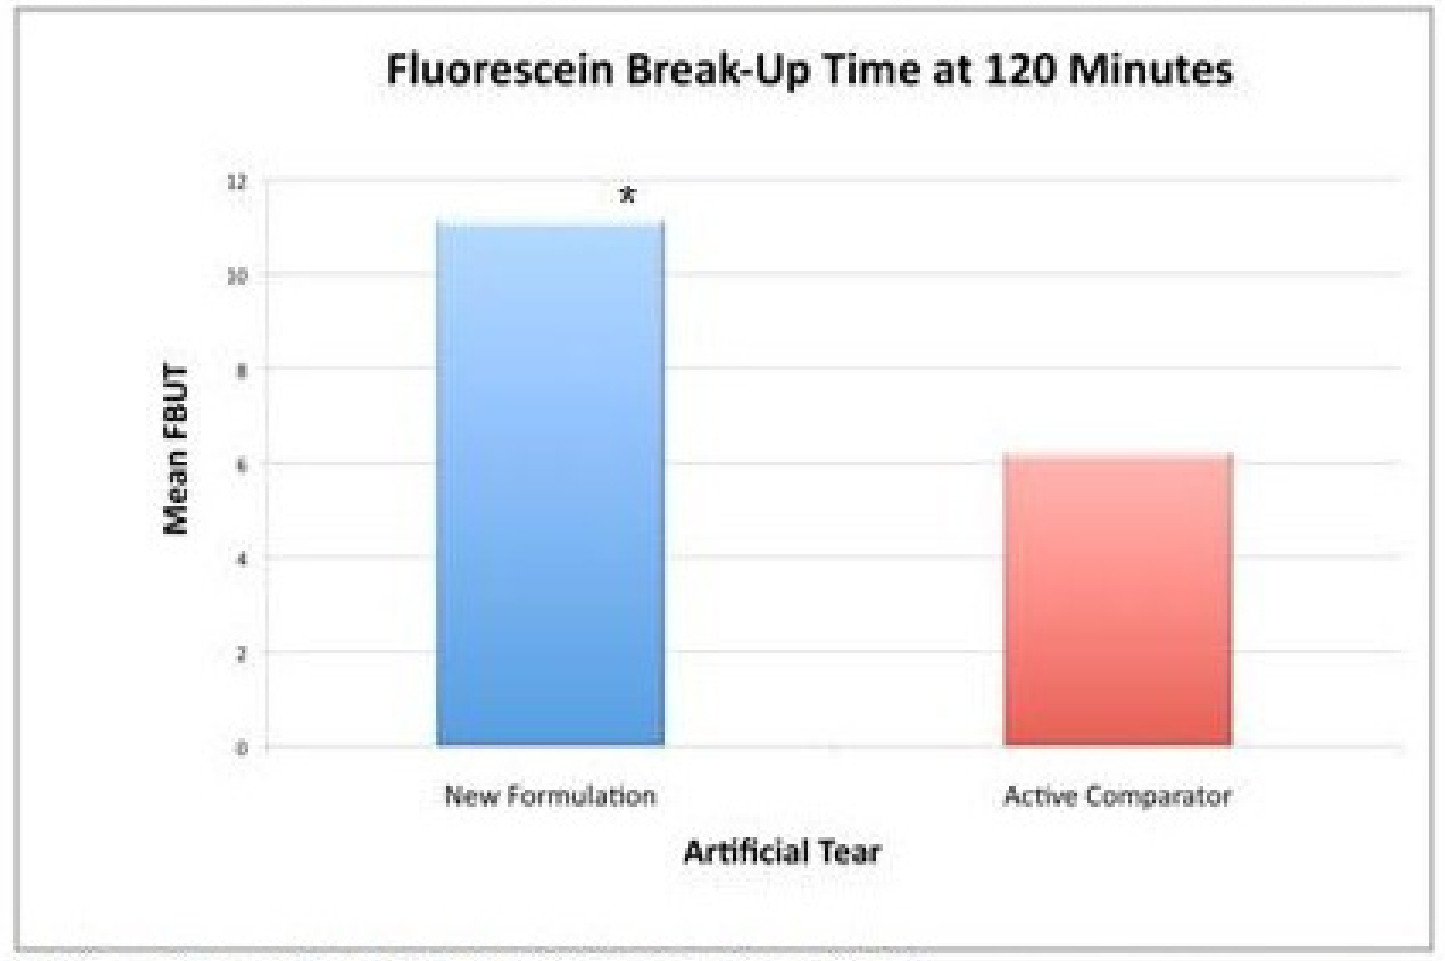
Fluorescein Break-Up Time at 120 Minutes
Mean FBUT
0
2
4
6
8
10
12
*
New Formulation
Active Comparator
Artificial Tear

Figure 2. Mean Fluorescein Break-Up Time (FBUT) at 120 minutes (N=16)
* The new formulation containing glycerin 1% had a FBUT 4.92 seconds longer than the active comparator at 120 minutes (p = 0.12)

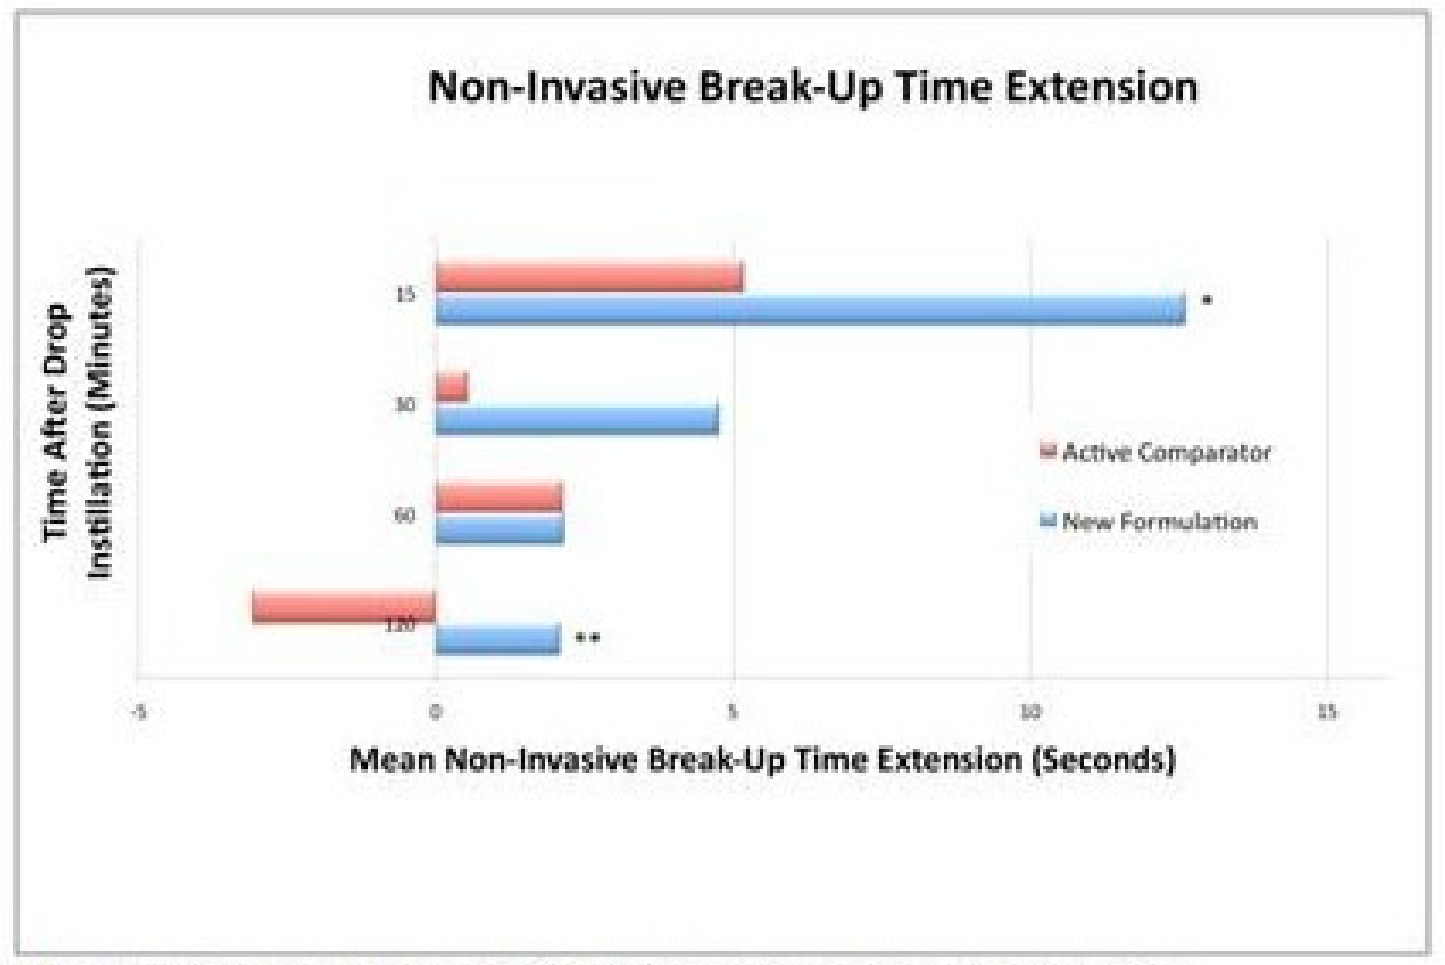
Non-Invasive Break-Up Time Extension
Time After Drop Instillation (Minutes)
15
30
60
120
*
**
Active Comparator
New Formulation
-5
0
5
10
15
Mean Non-Invasive Break-Up Time Extension (Seconds)

Figure 3. Mean Non-Invasive Break-Up Time (NIBUT) Extension with outliers removed (n=13)
* The new formulation containing glycerin 1% extended mean NIBUT at 15 minutes (p=0.08)
**The new formulation containing glycerin 1% had a mean NIBUT significantly longer than the active comparator at 120 minutes (p = 0.03)

RESULTS

The new, specifically engineered, formulation containing glycerin 1% extended NIBUT by 14.67 seconds at 15 minutes (p = 0.05) compared to pre-instillation NIBUT (Figure 1). The active comparator, which is a market leading product, extended NIBUT by 7.47 seconds at the same time point (p = 0.34). The new formulation of glycerin had a FBUT 4.92 seconds longer than the active comparator at 120 minutes (p = 0.12) (Figure 2).

Prior to unmasking, the principal investigator removed all subjects with TFBUT ≥ 30 seconds at baseline for a secondary data analysis. With outliers removed (n=13), mean NIBUT extension at 15 minutes for the new formulation was at 12.61 seconds (p = 0.08) (Figure 3). Mean NIBUT extension for the active comparator at the same time point was 5.17 seconds (p = 0.32). The difference between the mean NIBUT for the new formulation and active comparator at 120 minutes was statistically significant (p = 0.03).

CONCLUSION

This study suggests glycerin 1% can be formulated to significantly prolong NIBUT at 15 minutes, and that protective activity from an artificial tear product for two hours after eye drop instillation is possible. A trend showing that the new glycerin 1% formulation acts longer than a market leading product at 120 minutes was seen. A new formulation of glycerin 1% is a promising therapy for dry eye disease.

REFERENCES

1. Ousler GW, Michaelson C, Christensen MT. An evaluation of tear film breakup time extension and ocular protection index scores among three marketed lubricant eye drops. Cornea. 2007 Sep;26(8):949-52.
2. Paugh JR, Nguyen AL, Huang P, Hwang JS. Retention and retention of effect of topical formulations in dry eye subjects. Optom Vis Sci. 2008 Sep;85(9):873-9.
3. Prabhasawat P, Tesavibul N, Kasetsuwan N. Performance profile of sodium hyaluronate in patients with lipid tear deficiency: randomised, double-blind, controlled, exploratory study. Br J Ophthalmol. 2007 Jan;91(1):47-50. Epub 2006 Sep 14.
4. Simmons PA, Vehige JG. Clinical performance of a mid-viscosity artificial tear for dry eye treatment. Cornea. 2007 Apr;26(3):294-302.

SUPPORT: **None**

CLINICAL TRIAL: **www.clinicaltrials.gov NCT00681265**

CORRESPONDING AUTHOR: **David M. Kleinman, MD, MBA dave@eyeontherapeutics.com**

COMMERCIAL RELATIONSHIPS

W.G. Gensheimer, None; D.M. Kleinman, Eyeon Therapeutics, Inc., I; Eyeon Therapeutics, Inc., E; Bausch and Lomb, Inc., C; Eyeon Therapeutics, Inc., P; M.O. Gonzalez, None; D. Sobti, None; A. Loxley, Particle Sciences, Inc., E; Particle Sciences, Inc., P; M. Mitchnick, Particle Sciences, Inc., I; Particle Sciences, Inc., E; Particle Sciences, Inc., P; J.V. Aquavella, None.

PARTICLE SCIENCES
DRUG DEVELOPMENT SERVICES